Measuring the Impact of Academic-Industry Partnership

Lauren Wall, Sara Moellering, Amanda Spratt, Morgan Cooper, Aubrey Nielsen, Thelma Tennant, Marie Bowden, Brad Stanford, Meredith Hightower, Zeena Shelal, Erica Mills
University of Chicago and AbbVie

BACKGROUND

As oncology clinical research becomes increasingly complex, sustainable success requires strategic, performance-driven partnerships between academic cancer centers and industry sponsors. Traditional sponsor–site relationships are often transactional and lack shared performance accountability, limiting long-term value and operational optimization. Establishing aligned goals, strategic priorities and measurable outcomes is essential to advancing trial efficiency, quality, and research impact.

OBJECTIVE

To develop a strategic, scalable partnership with AbbVie extending beyond individual studies to one focused on broadening access to novel therapies to patients through a collaborative model supported by jointly defined key performance indicators (KPIs). The goal was to improve study execution, and long-term portfolio expansion while demonstrating institutional readiness for complex and early-phase trials.

SOLUTIONS IMPLEMENTED

Through structured governance and ongoing collaboration, the University of Chicago Medicine Comprehensive Cancer Center (UCCCC) and AbbVie Oncology co-developed a Scientific Alliance focused on a KPI framework and acceleration methodologies. To support these metrics, several operational enhancements were implemented, including structured meetings, pilot of an Electronic Health Record to Electronic Data Capture (EHR-to-EDC) technology platform to streamline data capture, defined formal communication and escalation pathways, implementation of a Master Informed Consent Form, standardization via a master "all programs" Confidential Disclosure Agreement, and establishment of a Master Clinical Trial Agreement and Master Rate Card. In parallel, a Rapid Activation Pilot was launched.

RESULTS

Prior to 2024, AbbVie-sponsored oncology trials at UCCCC totaled 0. Since expanding the strategic partnership, the collaboration has supported 10 lead Principal Investigators and enabled activation of 12 new studies (60% first-in-human), with 5 additional studies in the pipeline. Over the last year, we reduced study start up timelines by 17% while positively contributing to patient enrollment with 32% representing underrepresented minority populations. Lastly, the Rapid Activation Pilot successfully achieved trial opening in less than 60 days.

Operational Wins

Implementation of Operational Partnership Pod	Identified Pilot Study for EHR to EDC	Implementation of Rapid Activation Pilot	2-Day Turnaround for ICF Revisions Review
Master ICF	Master CDA	Master CSA	Rate Card

Key Performance Metrics

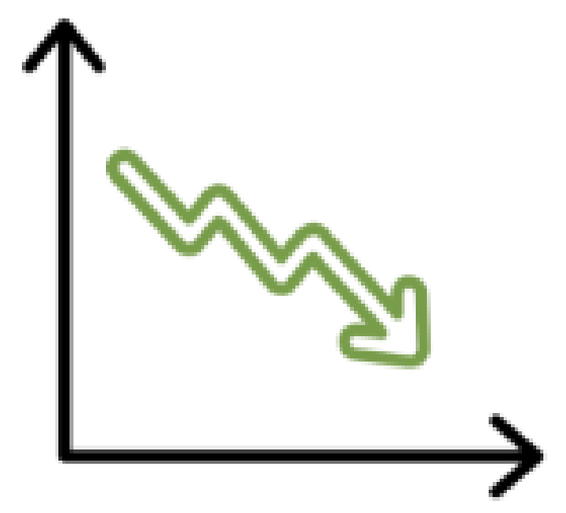

17%

Decrease in Activation Times

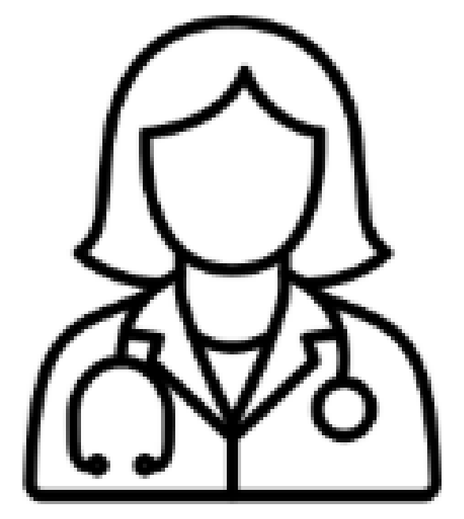

10

Unique PI's Involved in AbbVie Studies

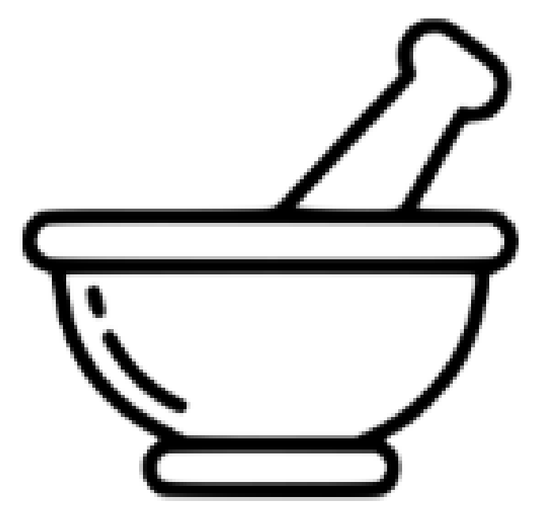

60%

Are First in Human Studies

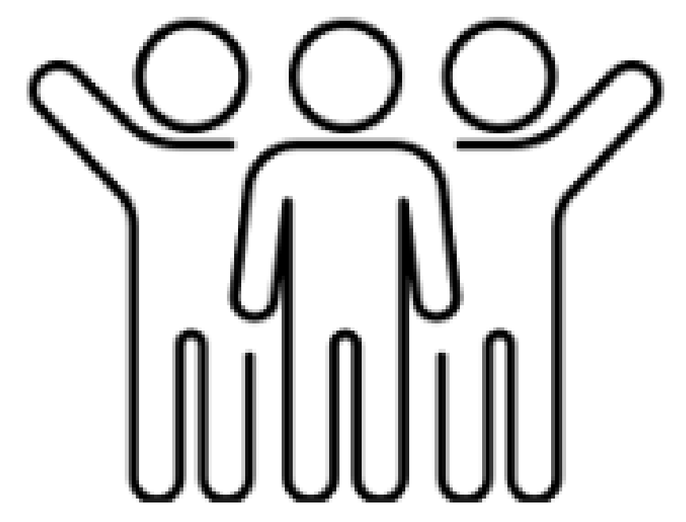

32%

Of Patients Enrolled are from Underrepresented Minorities

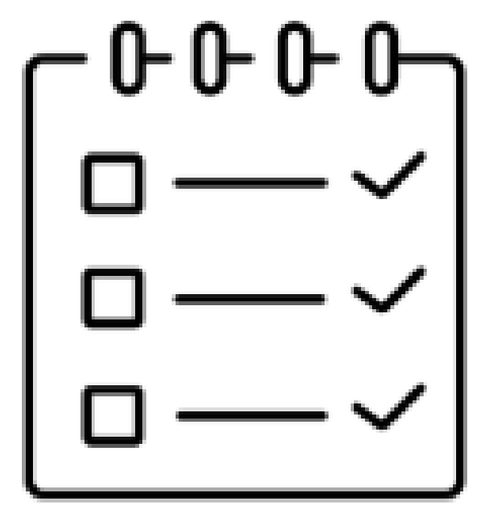

12

New Studies Opened since 2024

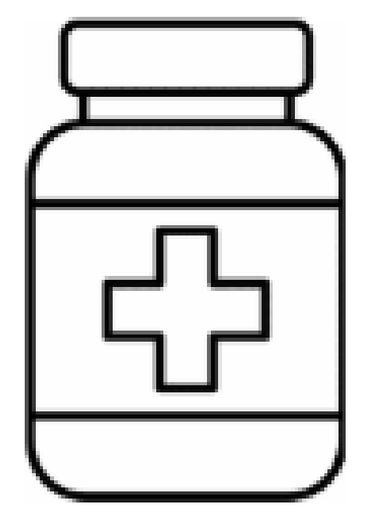

21

Patients Enrolled

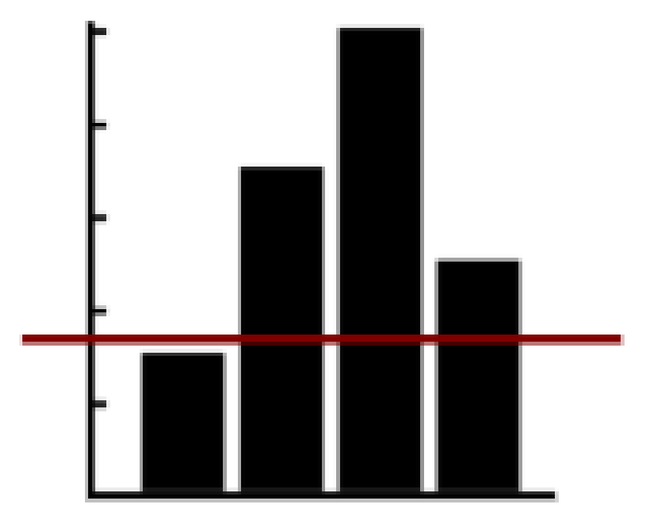

Establishing Baseline Activation Timeline to Better Identify Operational Efficiencies

LESSON LEARNED

This strategic partnership demonstrates that KPI-driven partnerships, coupled with targeted operational innovation, can create scalable, high-value industry collaborations that enhance efficiency, expand research access, and support long-term institutional and sponsor success.

Key lessons included:

- The importance of early alignment on expectations and success metrics
- The value of shared governance and regular performance check-ins
- The role of operational standardization in reducing administrative burden

Additionally, transparent communication and mutual trust were critical to enabling rapid problem-solving.

FUTURE DIRECTIONS

This partnership has been successful in increasing both efficiencies, access to AbbVie sponsored research, and we hope to duplicate this model with select strategic industry partners. Future directions for our AbbVie partnership include continuous improvement of clinical trial activation timeliness, addressing issues with trials coordinated via clinical research organizations (CROs) and ensuring that these trials follow the same established strategic partner pathways and KPIs.

AT THE FOREFRONT UChicago Medicine Comprehensive Cancer Center

abbvie